Intravenous Delivery of AAV Gene Therapy for the Treatment of SOD1-ALS Provides Broad SOD1 Lowering in NHP

Michael D. Grannan, Tyler Moyer, Margaret R. Dalrymple, Ambreen Sayed-Zahid, Xiao Ren, Chiso Egbuchulam, Amy Bruce, Anupriya Kulkarni, Wenya Yang, Joe Clement, Jeyashree Natasan, Dhiral Shah, Ruohong Zhou, Dillon Kavanagh, Kyle Grant, Jeffrey Thompson, Nilesh Pande, Beth Mellen, Naomi Muse, Crystal Taveras-Cui, Vikram Arora, Chanchal Sadhu, Dinah W.Y. Sah, Mathieu Nonnenmacher, Rajeev Sivasankaran, Todd Carter

Voyager Therapeutics Inc., Lexington, MA, USA

voyager therapeutics

SUMMARY

- Mutations in superoxide dismutase 1 (SOD1) are responsible for a toxic gain of function and cause SOD1 ALS
- Tofersen is an intrathecally delivered SOD1 ASO approved for the treatment of SOD1 ALS (USA and Europe), validating SOD1 reduction as a therapeutic target
- Using a capsid suitable for IV delivery in rodents and the same potent RNAi transgene used in VY9323, we have demonstrated robust knockdown of SOD1 in all levels of the spinal cord resulting in significant improvements in motor performance and survival in a mouse model of SOD1 ALS
- In primates, VY9323, an AAV gene therapy combining a potent pri-amiRNA against SOD1 with an intravenous (IV)-delivered, blood-brain barrier-penetrant TRACER™ capsid allowing for one time administration, provided substantial vector genome delivery to the spinal cord and motor cortex
- An IND submission is expected in mid-2025

INTRODUCTION

More than 180 mutations in the SOD1 gene have been linked to human disease and result in the formation of toxic aggregates that impair multiple cell functions, and result in dysfunction and degeneration of motor neurons. An intravenously administered therapy that utilizes CNS vasculature to deliver a therapeutic that reduces SOD1 may offer the opportunity for maximal therapeutic benefit. In the studies outlined here, we combined a highly potent siRNA against SOD1 with blood-brain barrier (BBB) penetrant capsids that allow for IV delivery in mouse and NHP. The results demonstrate that the combination of a potent SOD1 RNAi transgene with a novel TRACER™ capsid produces significant reduction of SOD1 mRNA in NHP in critical spinal cord and brain regions impacted in ALS and support its continued development and advancement into the clinic.

Figure 1. SOD1 Knockdown Rationale and SOD1 siRNA Selection

Lowering of SOD1 Provides Therapeutic Benefit in Humans:

- Partial lowering of SOD1 provides potential therapeutic benefit and is well-tolerated in animal models, providing a useful tool to test our therapeutic modality
- Reduction of SOD1 by tofersen results in NfL reduction and potential therapeutic benefit in humans. NfL is a marker of neuron and axonal degeneration and is prognostic for survival and function in ALS

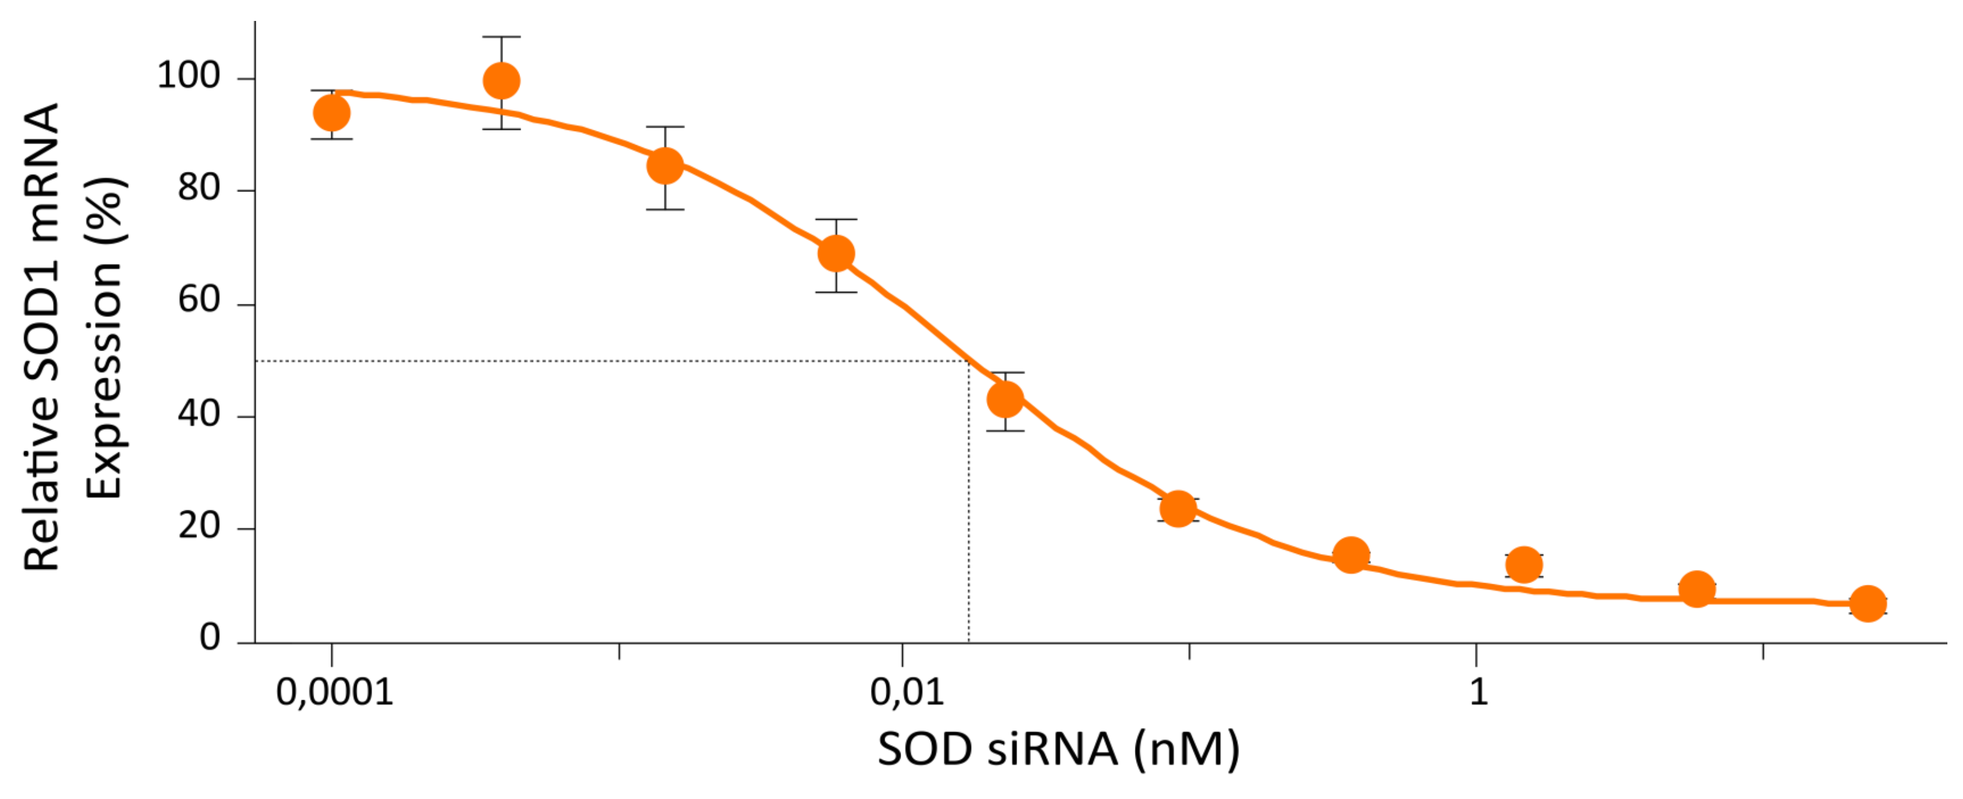

Relative SOD1 mRNA Expression (%)
SOD siRNA (nM)

SOD1 mRNA silencing *in vitro* by the potent siRNA utilized in VY9323. Sequences selectively targeting SOD1 were designed, synthesized and evaluated in HeLa cells. 24 hr after transfection of 100 pM SOD1 siRNA, cells were harvested, and SOD1 and GAPDH mRNA were quantified by RT-qPCR. Dose response curve of the lead candidate is depicted. Error bars indicate SD.

SOD1 G93A Efficacy Study

Mice:	~56-day old B6SJL-Tg(SOD1*G93A)1Gur/J (male and female)
Vector:	VOY101.SOD1 miRNA
Dose:	100µl, IV administration 2e12, 6.3e12, and 2e13 vg/kg
Endpoints:	Motor performance, body weight, survival, SOD1 knockdown in spinal cord

Figure 2. Broad Vector Genome Biodistribution and SOD1 Knockdown in Mouse Spinal Cord

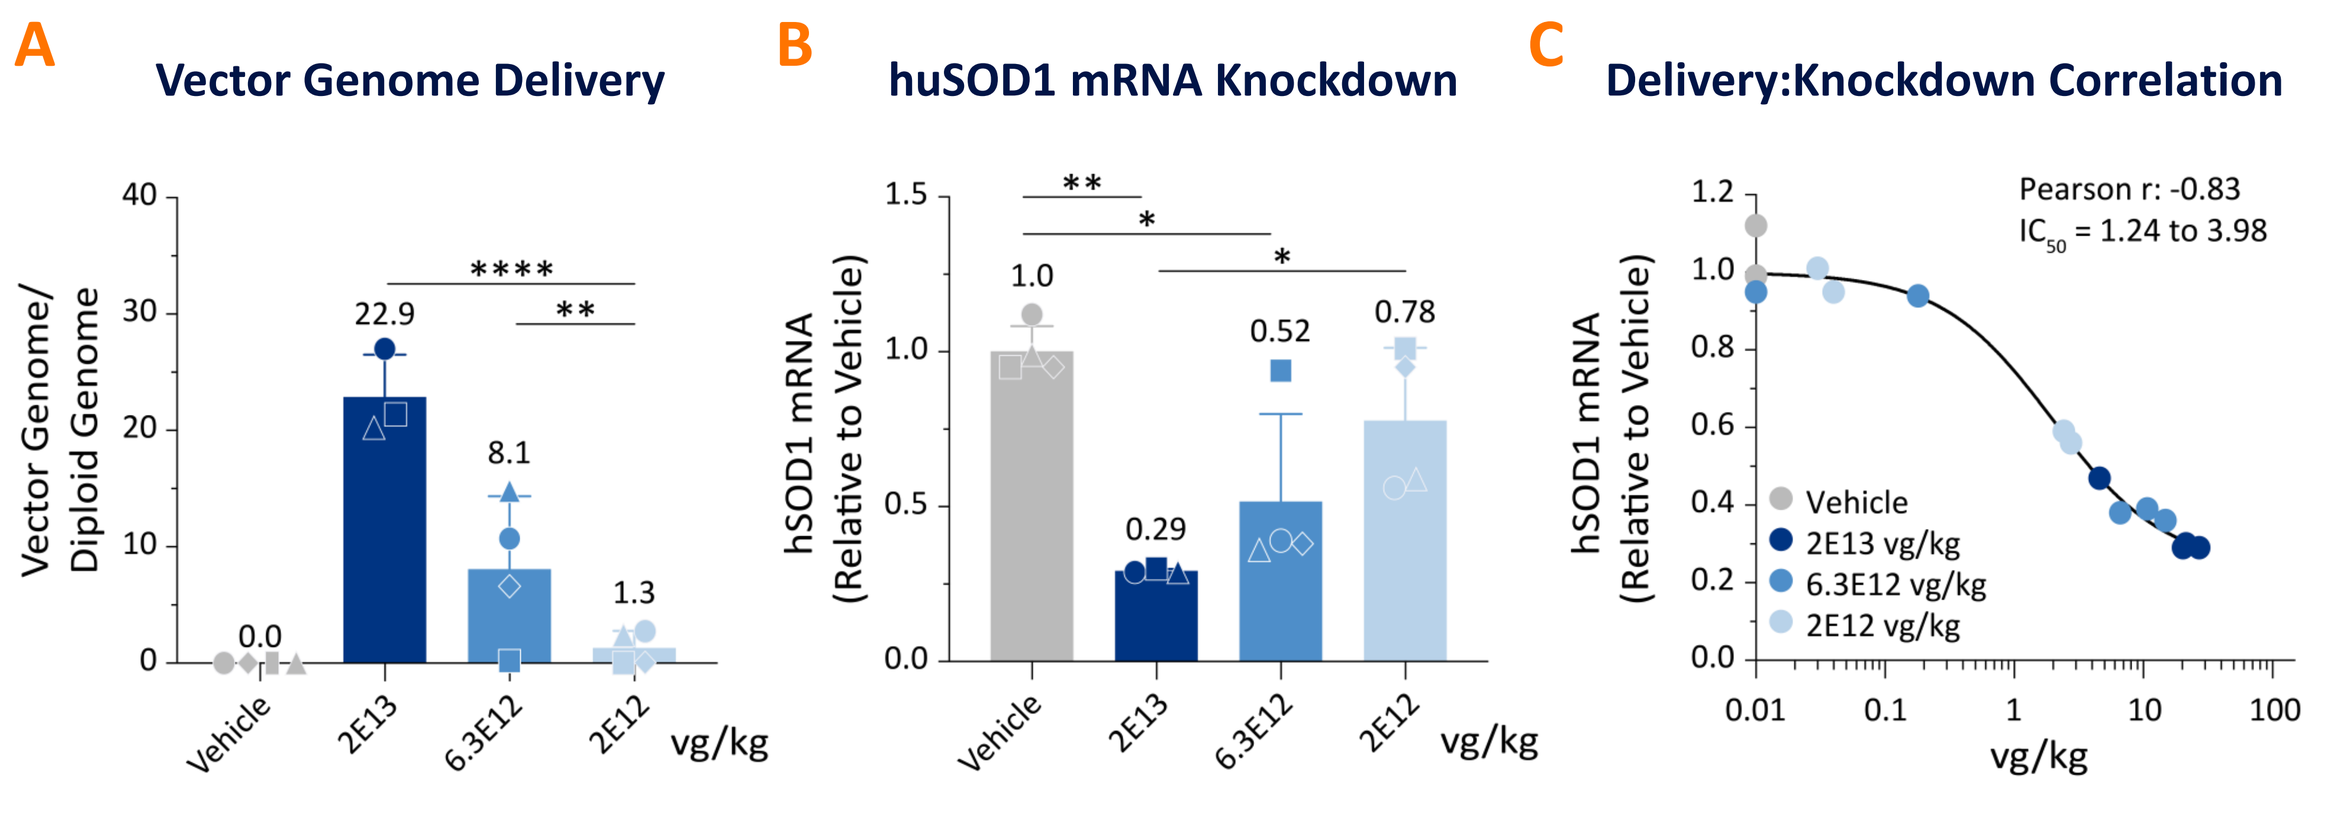

A Vector Genome Delivery
B huSOD1 mRNA Knockdown
C Delivery:Knockdown Correlation
Pearson r: -0.83
IC_{50} = 1.24 to 3.98

Vector genome distribution and hSOD1 knockdown in the cervical, thoracic, and lumbar spinal cord of G93A mice 32 days following AAV administration (representative cervical data shown here). (**A**) Vector genome distribution was analyzed using a multiplex ddPCR assay. (**B**) hSOD1 expression measured using multiplexed RT-qPCR (**C**) Correlation of vector genome to hSOD1 knockdown in the mouse spinal cord. *p<0.05, **0.01, ***0.001, 1-way ANOVA with Tukey's Multiple Comparisons.

Figure 3. Increase in Survival in G93A Mice

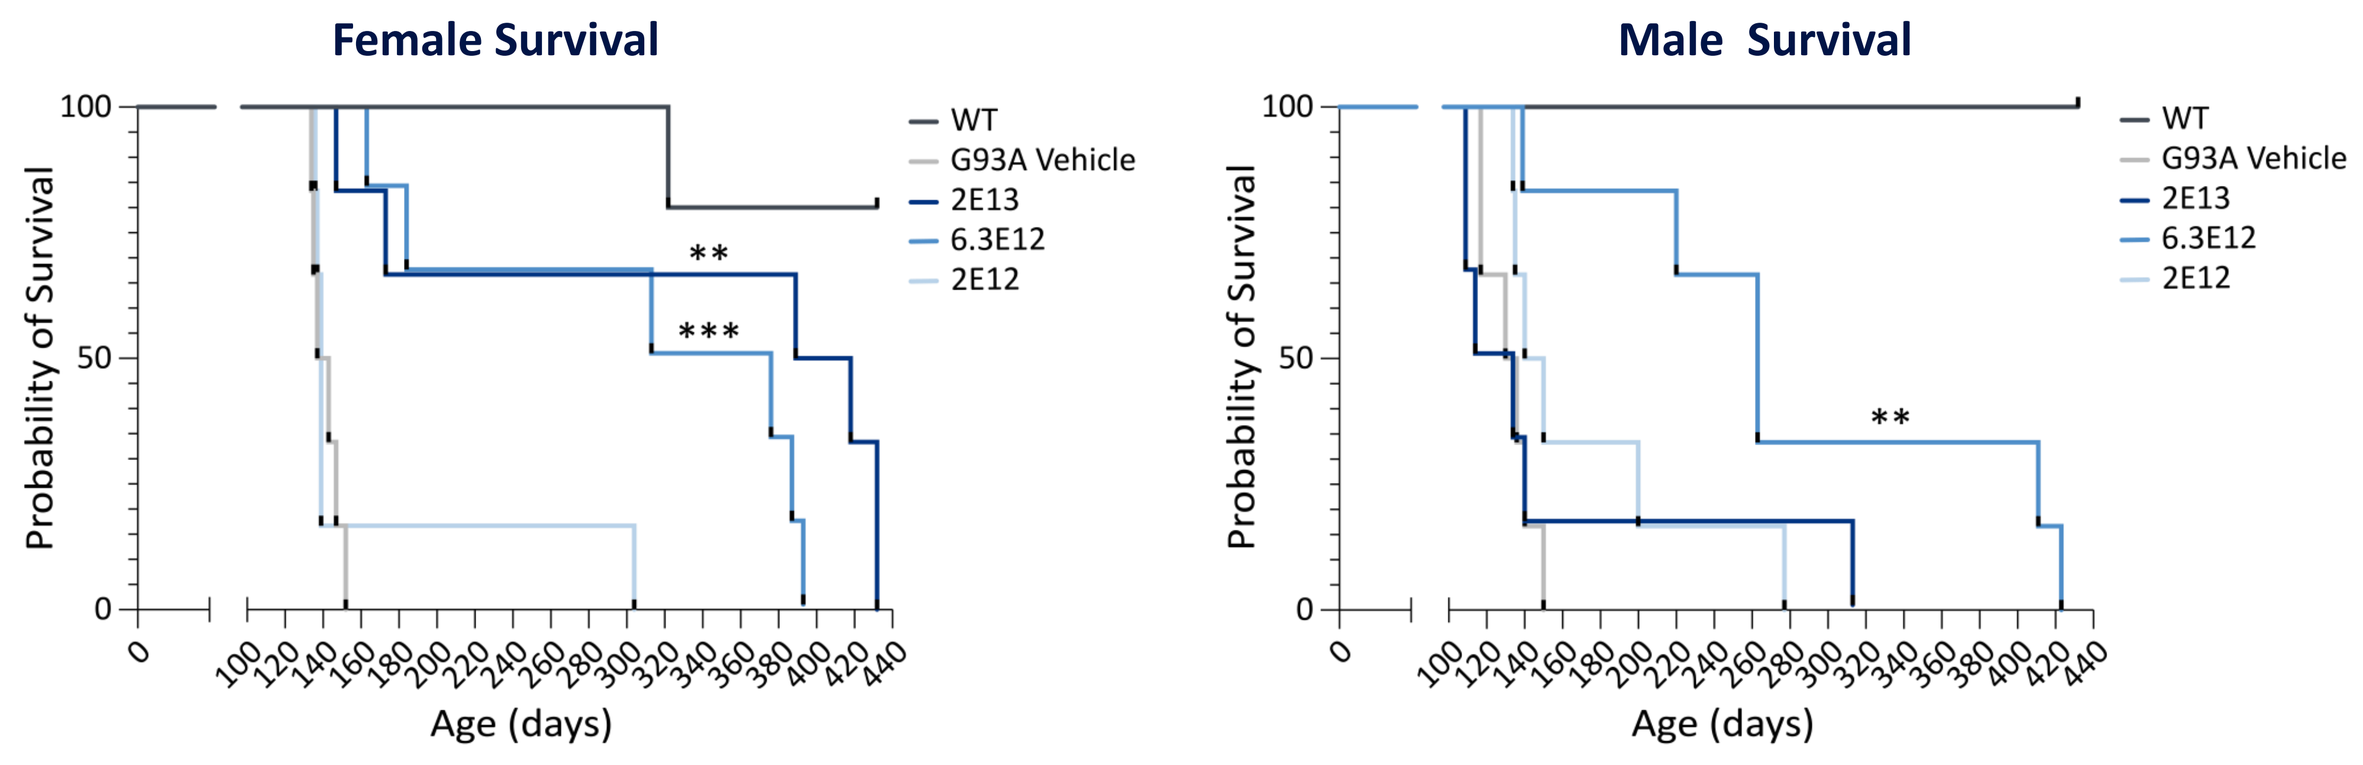

Female Survival
Male Survival
Probability of Survival
Age (days)
WT
G93A Vehicle
2E13
6.3E12
2E12

Female	Median Survival (days)	Improvement From Vehicle (days)	Significance (p=)
G93A Vehicle	140	N/A	N/A
2E12 vg/kg	139	-1	ns
6.3E12 vg/kg	345	205	0.0005
2E13 vg/kg	404	264	0.0022

Male	Median Survival (days)	Improvement From Vehicle (days)	Significance (p=)
G93A Vehicle	133	N/A	N/A
2E12 vg/kg	145	12	ns
6.3E12 vg/kg	263	133	0.003
2E13 vg/kg	124	-9	ns

Reduction of spinal cord SOD1 results in increased survival in G93A mice. Median survival was 140 days for vehicle treated female mice and 139 (n.s.) days for low dose animals. Median survival for moderate and high doses were 345 and 404 days, respectively. Median survival was 133 days for vehicle treated males, 124 (n.s.) and 145 (n.s.) days for the high and low dose, respectively, and 263 (**p = 0.003) days for the mid dose group. Statistical analysis was performed using log-rank (Mantel-Cox) test.

AAV9-based TRACER™ screen identifies cross-species, BBB-penetrant capsids with conserved receptor in mouse, NHP, and human

Figure 4. VY9323 Capsid Discovery Workflow and Resulting Improvements in CNS Targeting

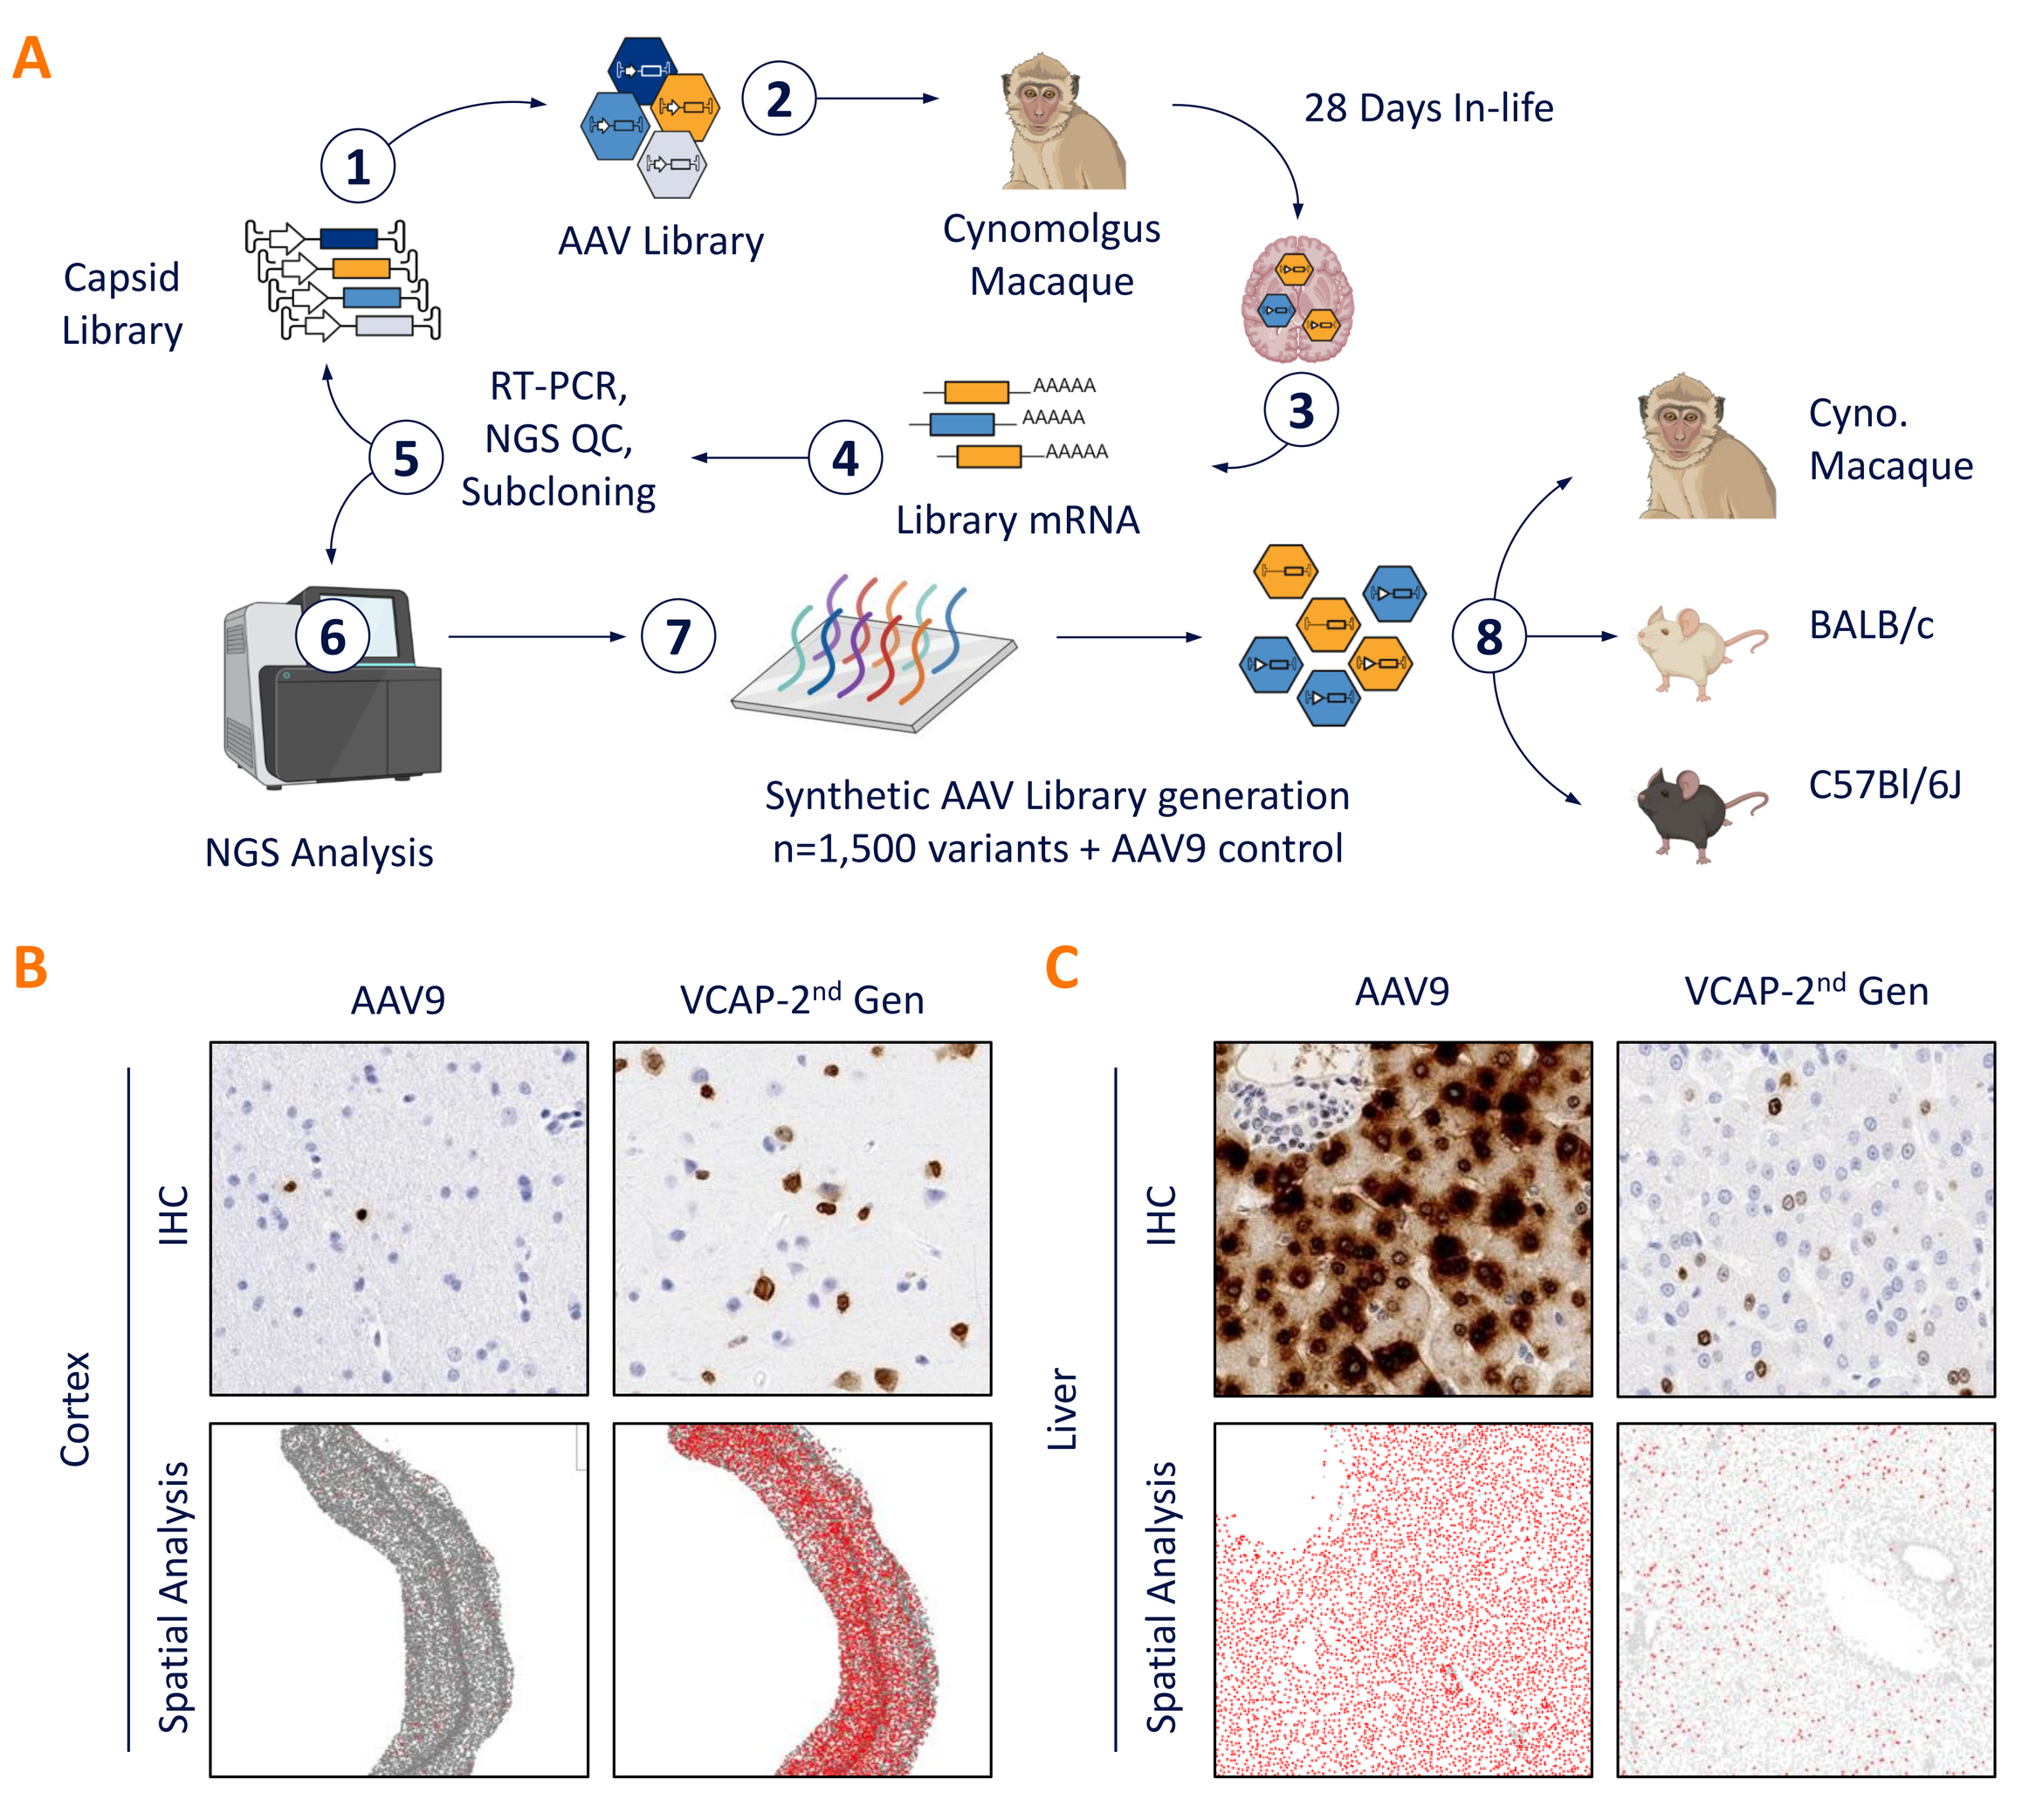

A
Capsid Library
AAV Library
Cynomolgus Macaque
28 Days In-life
Library mRNA
RT-PCR, NGS QC, Subcloning
NGS Analysis
Synthetic AAV Library generation n=1,500 variants + AAV9 control
Cyno. Macaque
BALB/c
C57Bl/6J
B
AAV9
VCAP-2nd Gen
Cortex
IHC
Spatial Analysis
C
AAV9
VCAP-2nd Gen
Liver
IHC
Spatial Analysis

(**A**) Design of TRACER-NHP directed evolution pipeline depicted. Second generation candidates resulting from fitness maturation were selected for increased CNS tropism and liver detargeting. (**B, C**) Expression of alternate tagged transgene used to illustrate improved transduction in cortex compared to AAV9, while detargeting the liver.

Figure 5. Identification of a Highly Conserved Brain Vascular Receptor, ALPL (Alkaline Phosphatase), Specifically Bound by the Cross-species VCAP 2nd Generation Capsid Utilized in VY9323

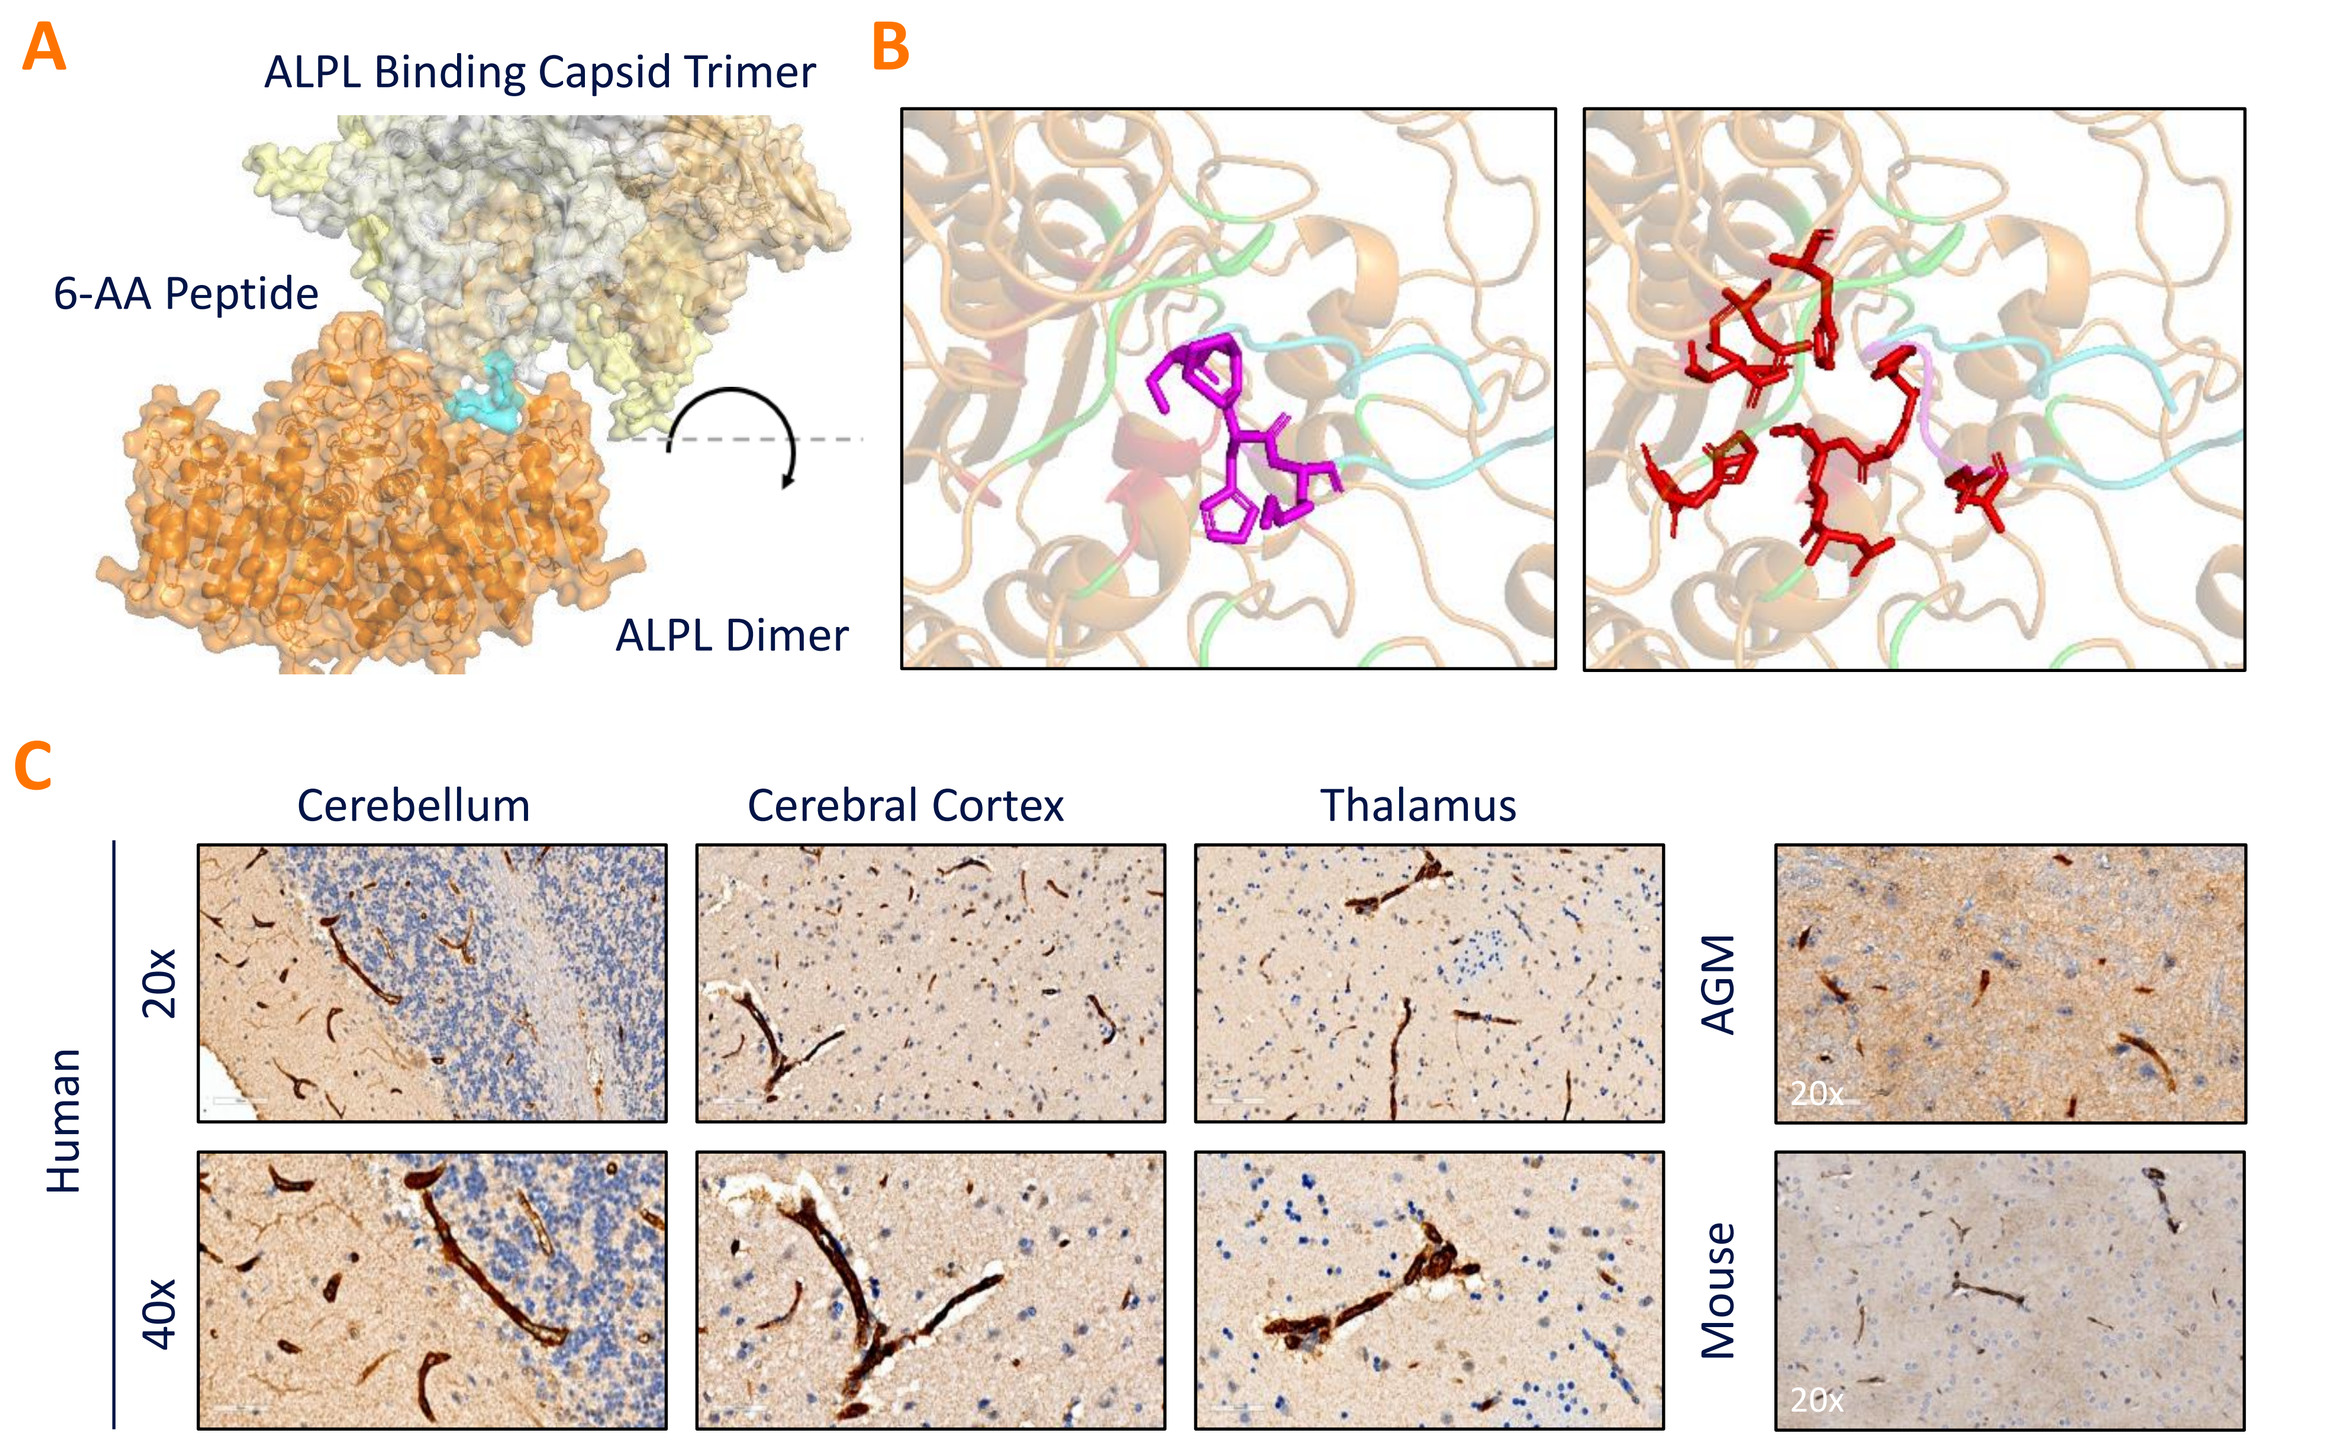

A
ALPL Binding Capsid Trimer
6-AA Peptide
ALPL Dimer
B
C
Cerebellum
Cerebral Cortex
Thalamus
Human
20x
40x
AGM
Mouse

(**A**) AlphaFold2-multimer predicted structure of ALPL dimer (orange) and an ALPL binding capsid VP3 trimer (white, yellow, wheat) with the VCAP 2nd generation 6-AA peptide shown in turquoise. (**B**) Top-down view of the ALPL active pocket bound to a VCAP 2nd generation peptide (turquoise) . Top panel: conserved residues on the peptide are shown in magenta, ALPL residues predicted to interact with VCAP 2nd generation are shown in green. Right panel: ALPL residues in contact with the conserved motif of the peptide are highlighted in red. (**C**) Detection of ALPL by immunohistochemistry (IHC) in brain sections from human, African green monkey and mouse.

SOD1 Pharmacology Study in NHP

NHP:	2-5yo Male & Female Cynomolgus monkey (Macaca fascicularis); ~4 kg
Vector:	VCAP-2nd Generation.SOD1 miRNA **(VY9323)**
Dose:	IV administration, 3e13 vg/kg
Endpoints:	Biodistribution, SOD1 reduction, tolerability

Figure 6. Broad Vector Genome Biodistribution, SOD1 Knockdown, and Motor Neuron Transduction in NHP Spinal Cord

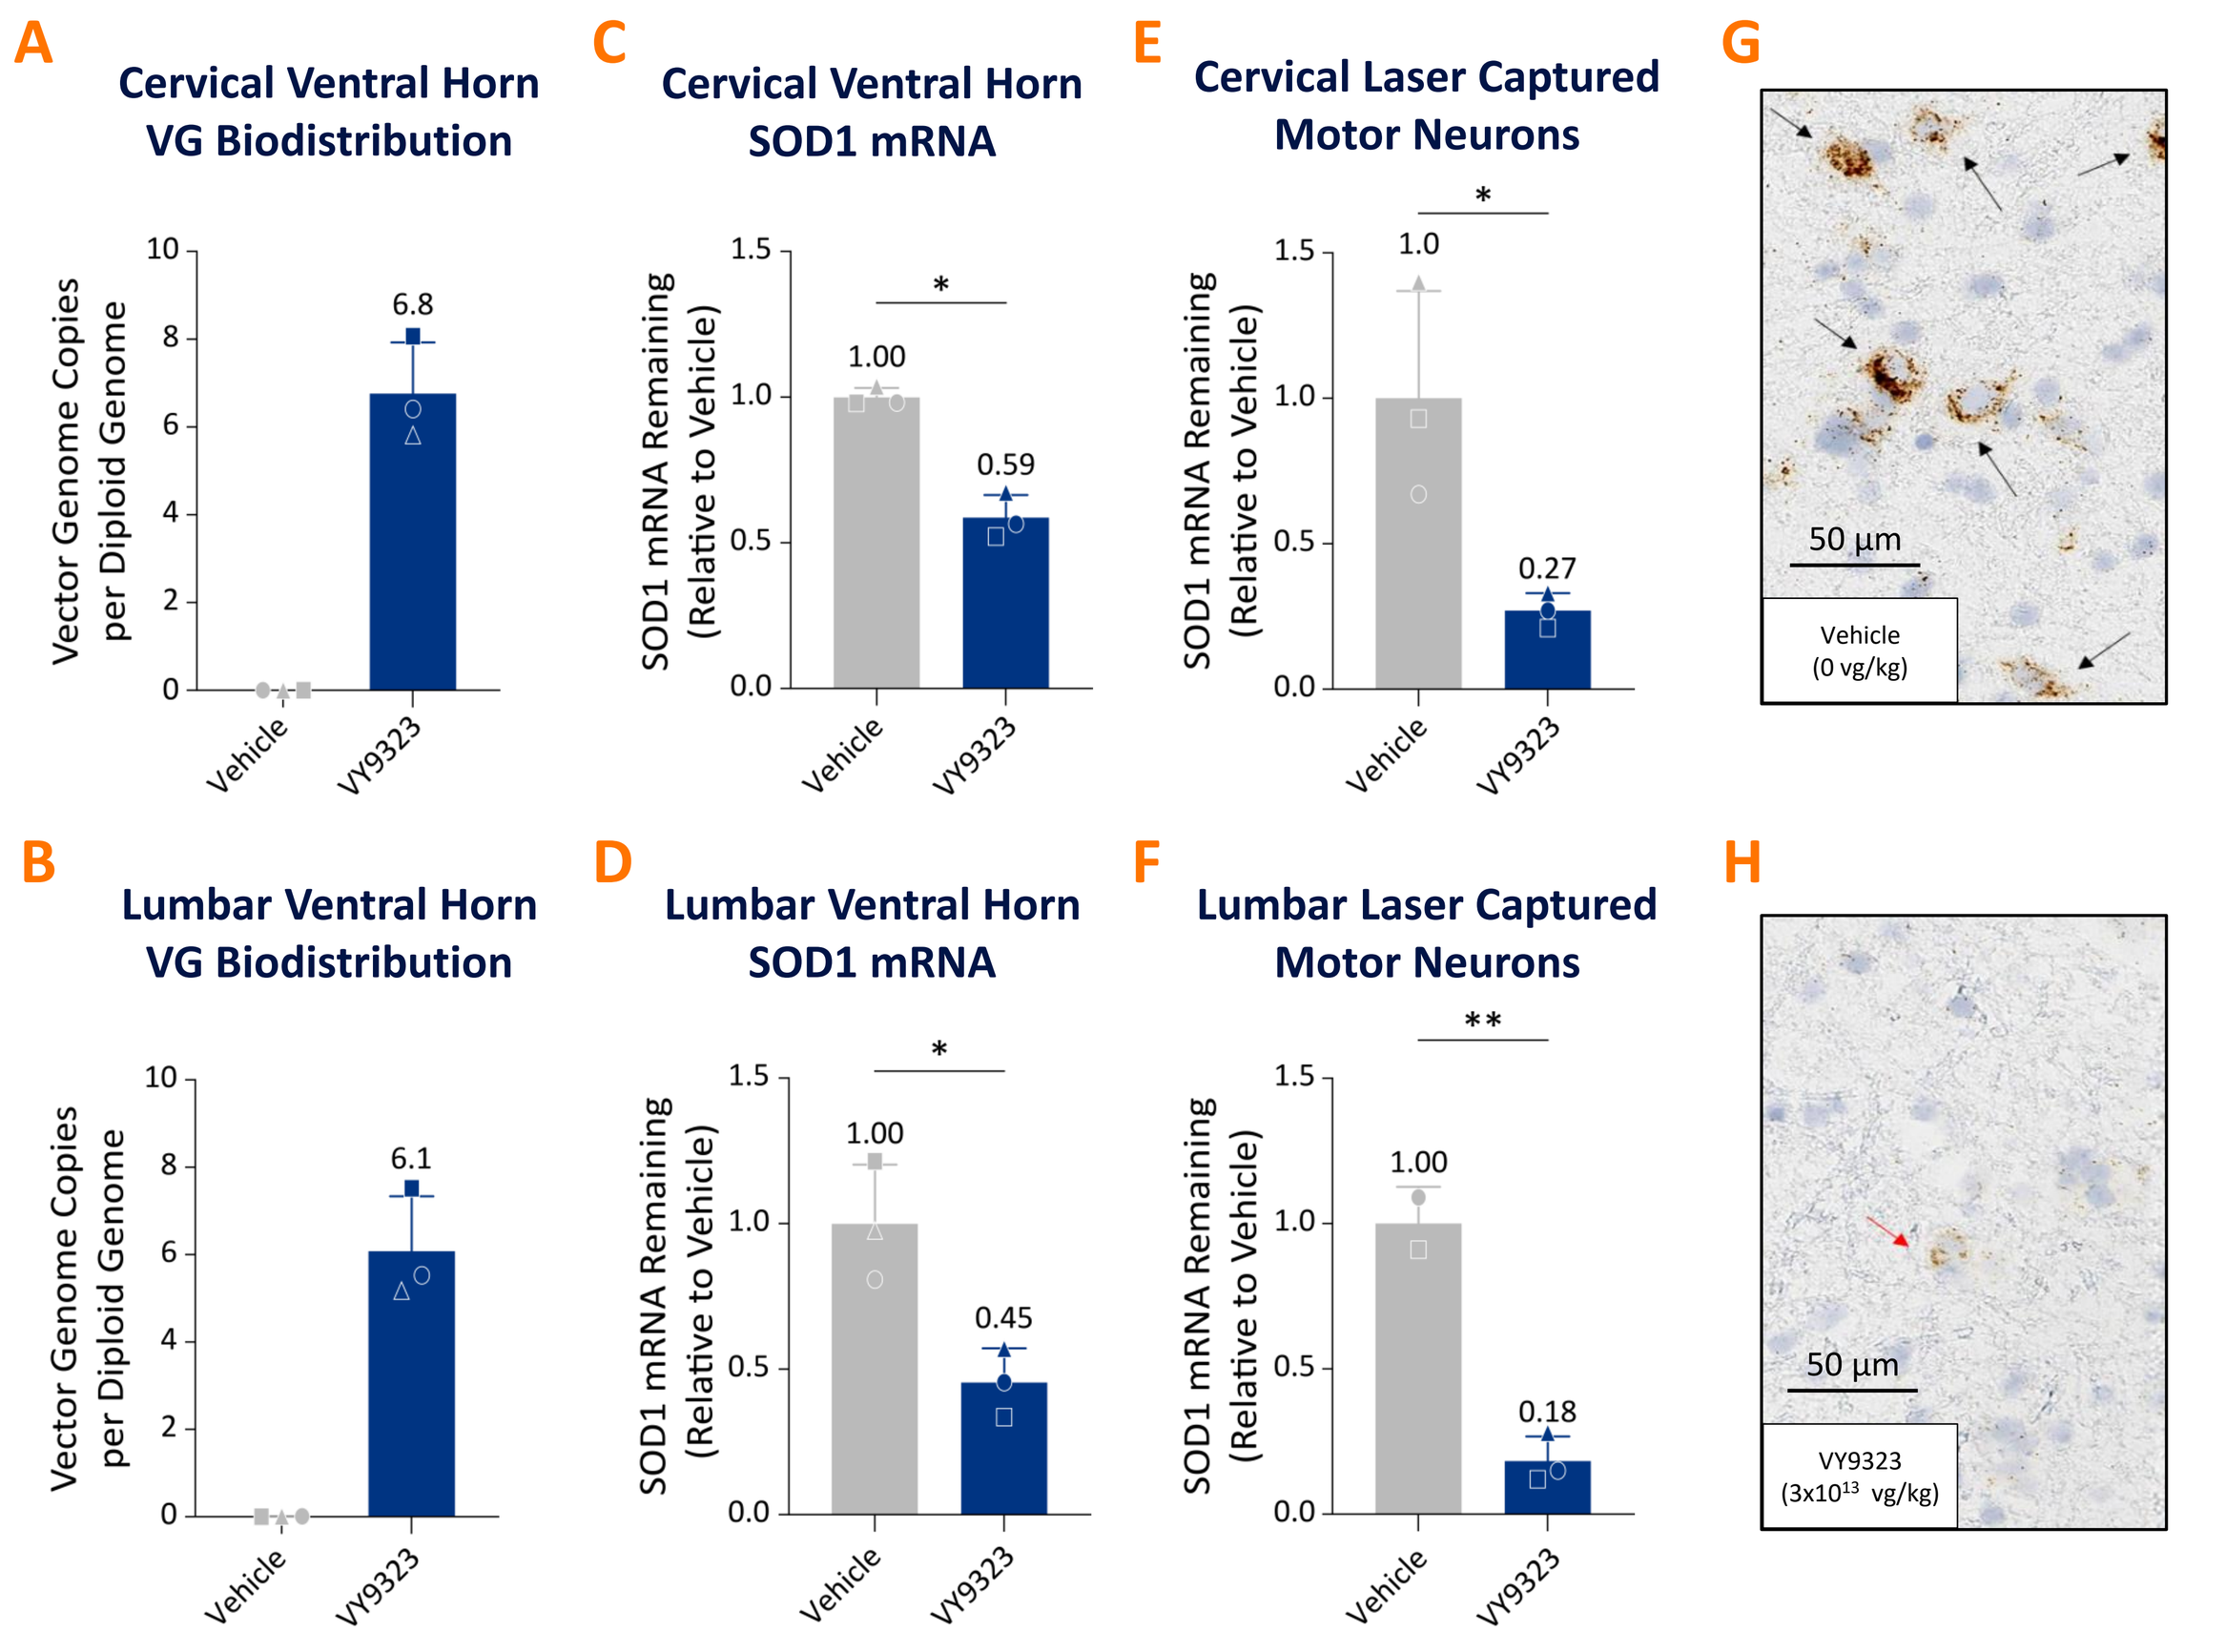

A Cervical Ventral Horn VG Biodistribution
B Lumbar Ventral Horn VG Biodistribution
C Cervical Ventral Horn SOD1 mRNA
D Lumbar Ventral Horn SOD1 mRNA
E Cervical Laser Captured Motor Neurons
F Lumbar Laser Captured Motor Neurons
G
H
Vector Genome Copies per Diploid Genome
SOD1 mRNA Remaining (Relative to Vehicle)
Vehicle
VY9323
50 µm
Vehicle (0 vg/kg)
VY9323 (3x10^13 vg/kg)

Intravenous administration of VY9323 in cynomolgus monkeys results in substantial vector genome delivery (**A**, **B**) in the cervical and lumbar spinal cord and produces significant SOD1 mRNA reduction in cervical and lumbar spinal cord ventral horn tissue punches (**C**, **D**) and laser captured motor neurons (**E**, **F**). VY9323 substantially reduces SOD1 mRNA in cervical spinal cord when evaluated using RNAscope (**G**, **H**). Black arrows represent cells expressing many copies of SOD1 mRNA, and red arrows indicate cells that express fewer copies. Automated detection of motor neuron transduction following delivery of a tagged payload and the VY9323 capsid determined 85-94% of neurons were transduced in cervical, thoracic, and lumbar spinal cord.

Figure 7. VY9323 Transduces Motor Neurons in Layer V of the Motor Cortex and Hypoglossal Nucleus, Regions Important for ALS Disease Progression

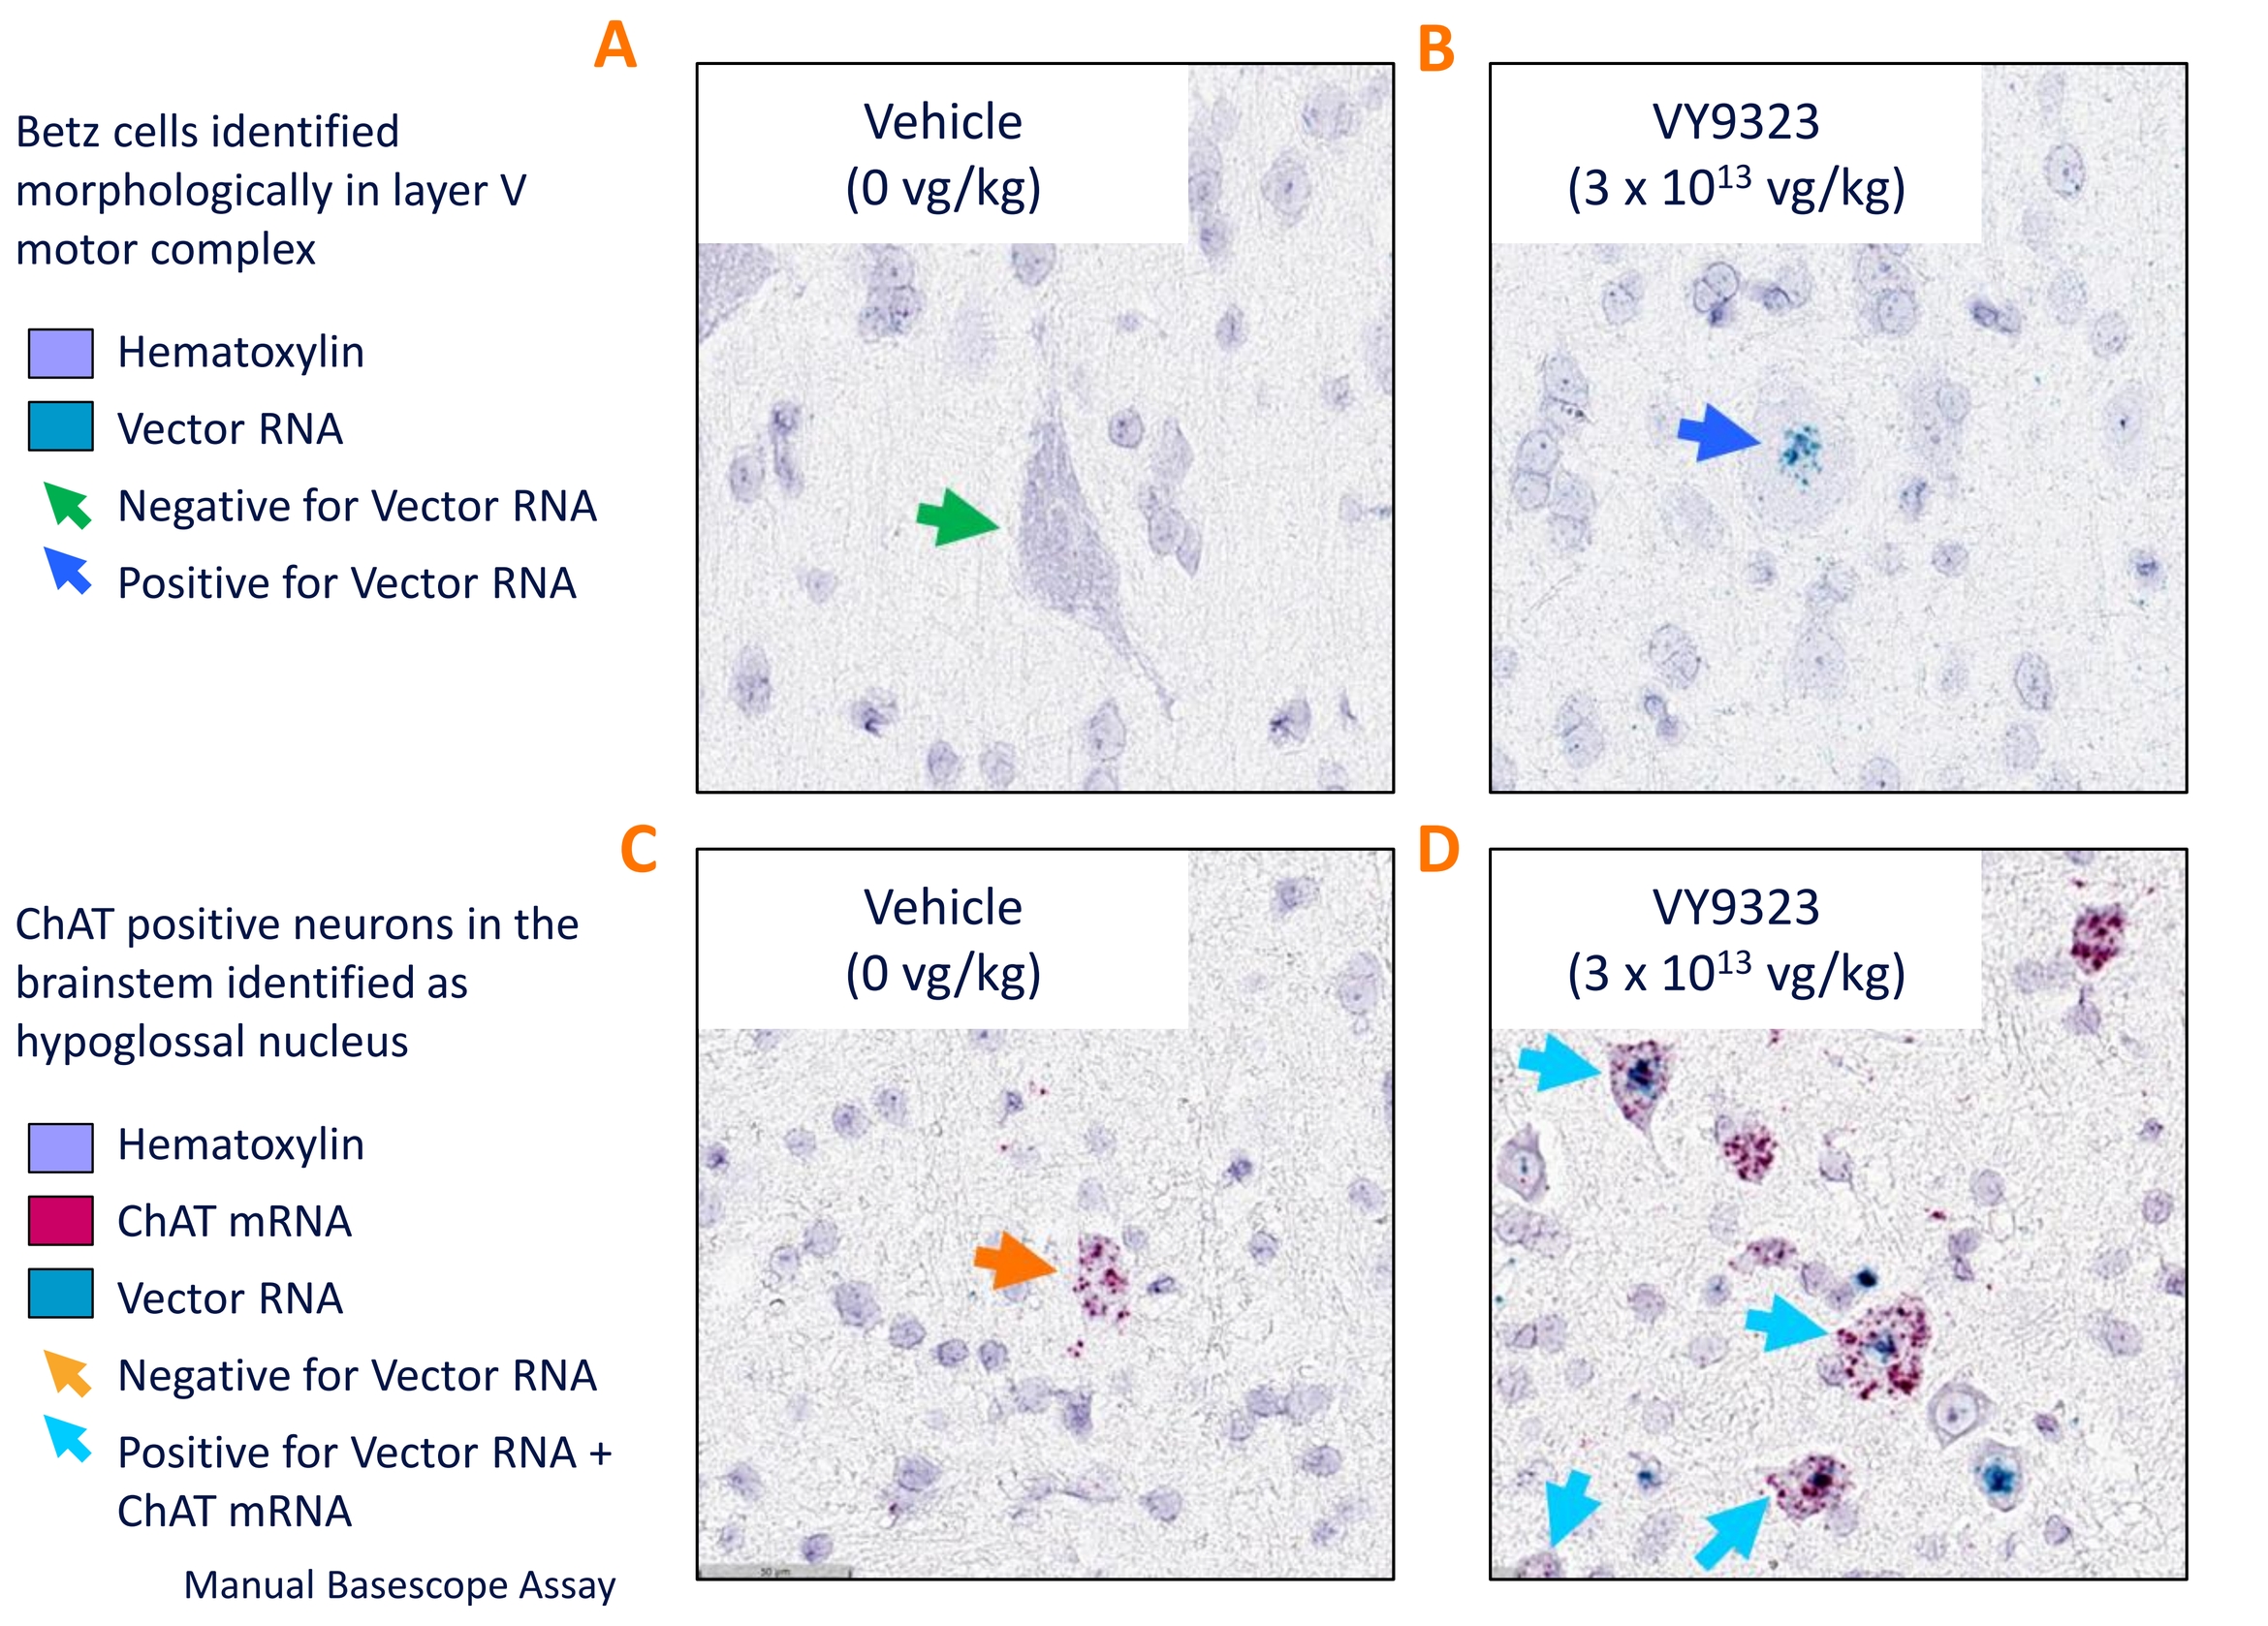

Betz cells identified morphologically in layer V motor complex
Hematoxylin
Vector RNA
Negative for Vector RNA
Positive for Vector RNA
ChAT positive neurons in the brainstem identified as hypoglossal nucleus
Hematoxylin
ChAT mRNA
Vector RNA
Negative for Vector RNA
Positive for Vector RNA + ChAT mRNA
Manual Basescope Assay
A Vehicle (0 vg/kg)
B VY9323 (3 x 10^13 vg/kg)
C Vehicle (0 vg/kg)
D VY9323 (3 x 10^13 vg/kg)

(**A**) Morphologically-identified motor neurons visualized in layer V of the motor cortex (green arrow). (**B**) VY9323 administration resulted in Betz cell transduction visualized utilizing BaseScope (blue arrow). (**C**) Cholinergic neurons in the hypoglossal nucleus of the brainstem are identified (orange arrow) and are transduced by VY9323 (**D**, blue arrows).

Figure 8. Enhanced Biodistribution in Motor Cortex and Decrease in Peripheral Tissues is Optimal for Efficacy and Safety

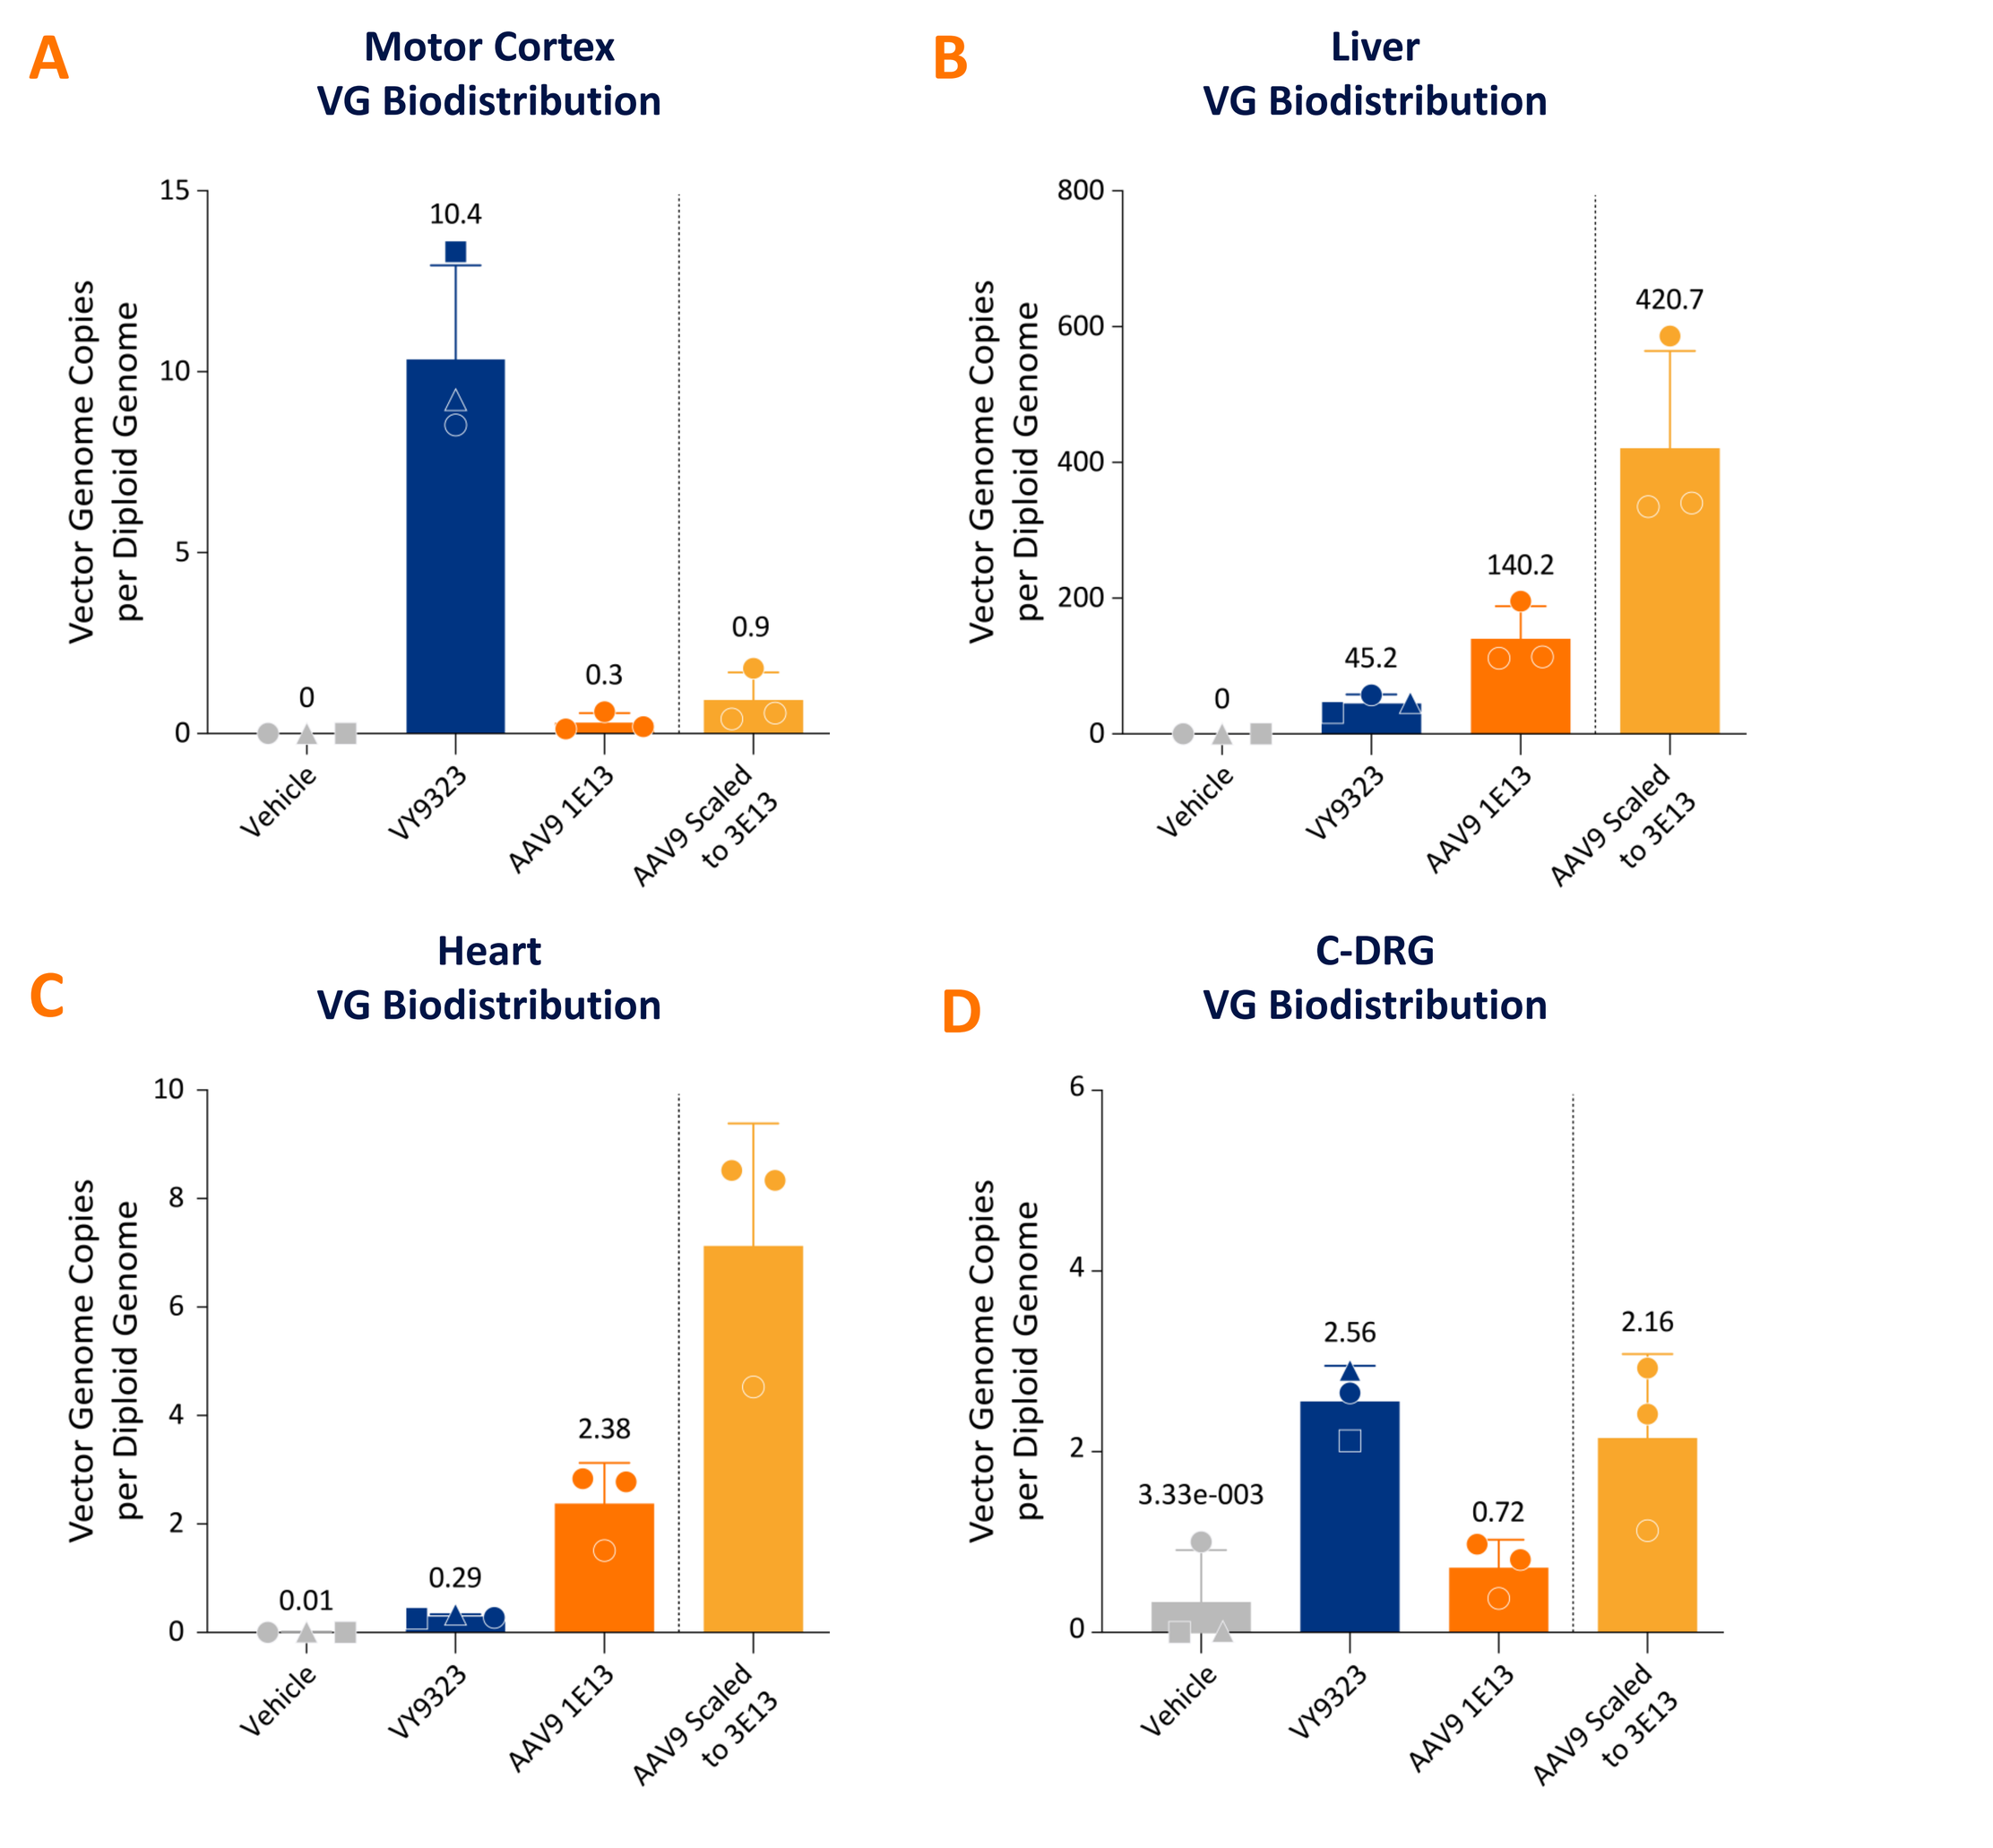

A Motor Cortex VG Biodistribution
B Liver VG Biodistribution
C Heart VG Biodistribution
D C-DRG VG Biodistribution
Vector Genome Copies per Diploid Genome
Vehicle
VY9323
AAV9 1E13
AAV9 Scaled to 3E13

(**A**) Intravenous administration of VY9323 in cynomolgus monkeys results in substantial vector genome delivery in the motor cortex while exhibiting peripheral tissue detargeting (**B**-**D**) compared to historical WT-AAV9 data expressing an alternative transgene at a 1E13 vg/kg dose scaled to 3E13 vg/kg. C-DRG = Cervical Dorsal Root Ganglia. Error bars represent standard deviation.

Figure 9. Clinical Chemistry and Hematology Data do not Present Meaningful Liabilities in NHP and Support Detargeting of Liver

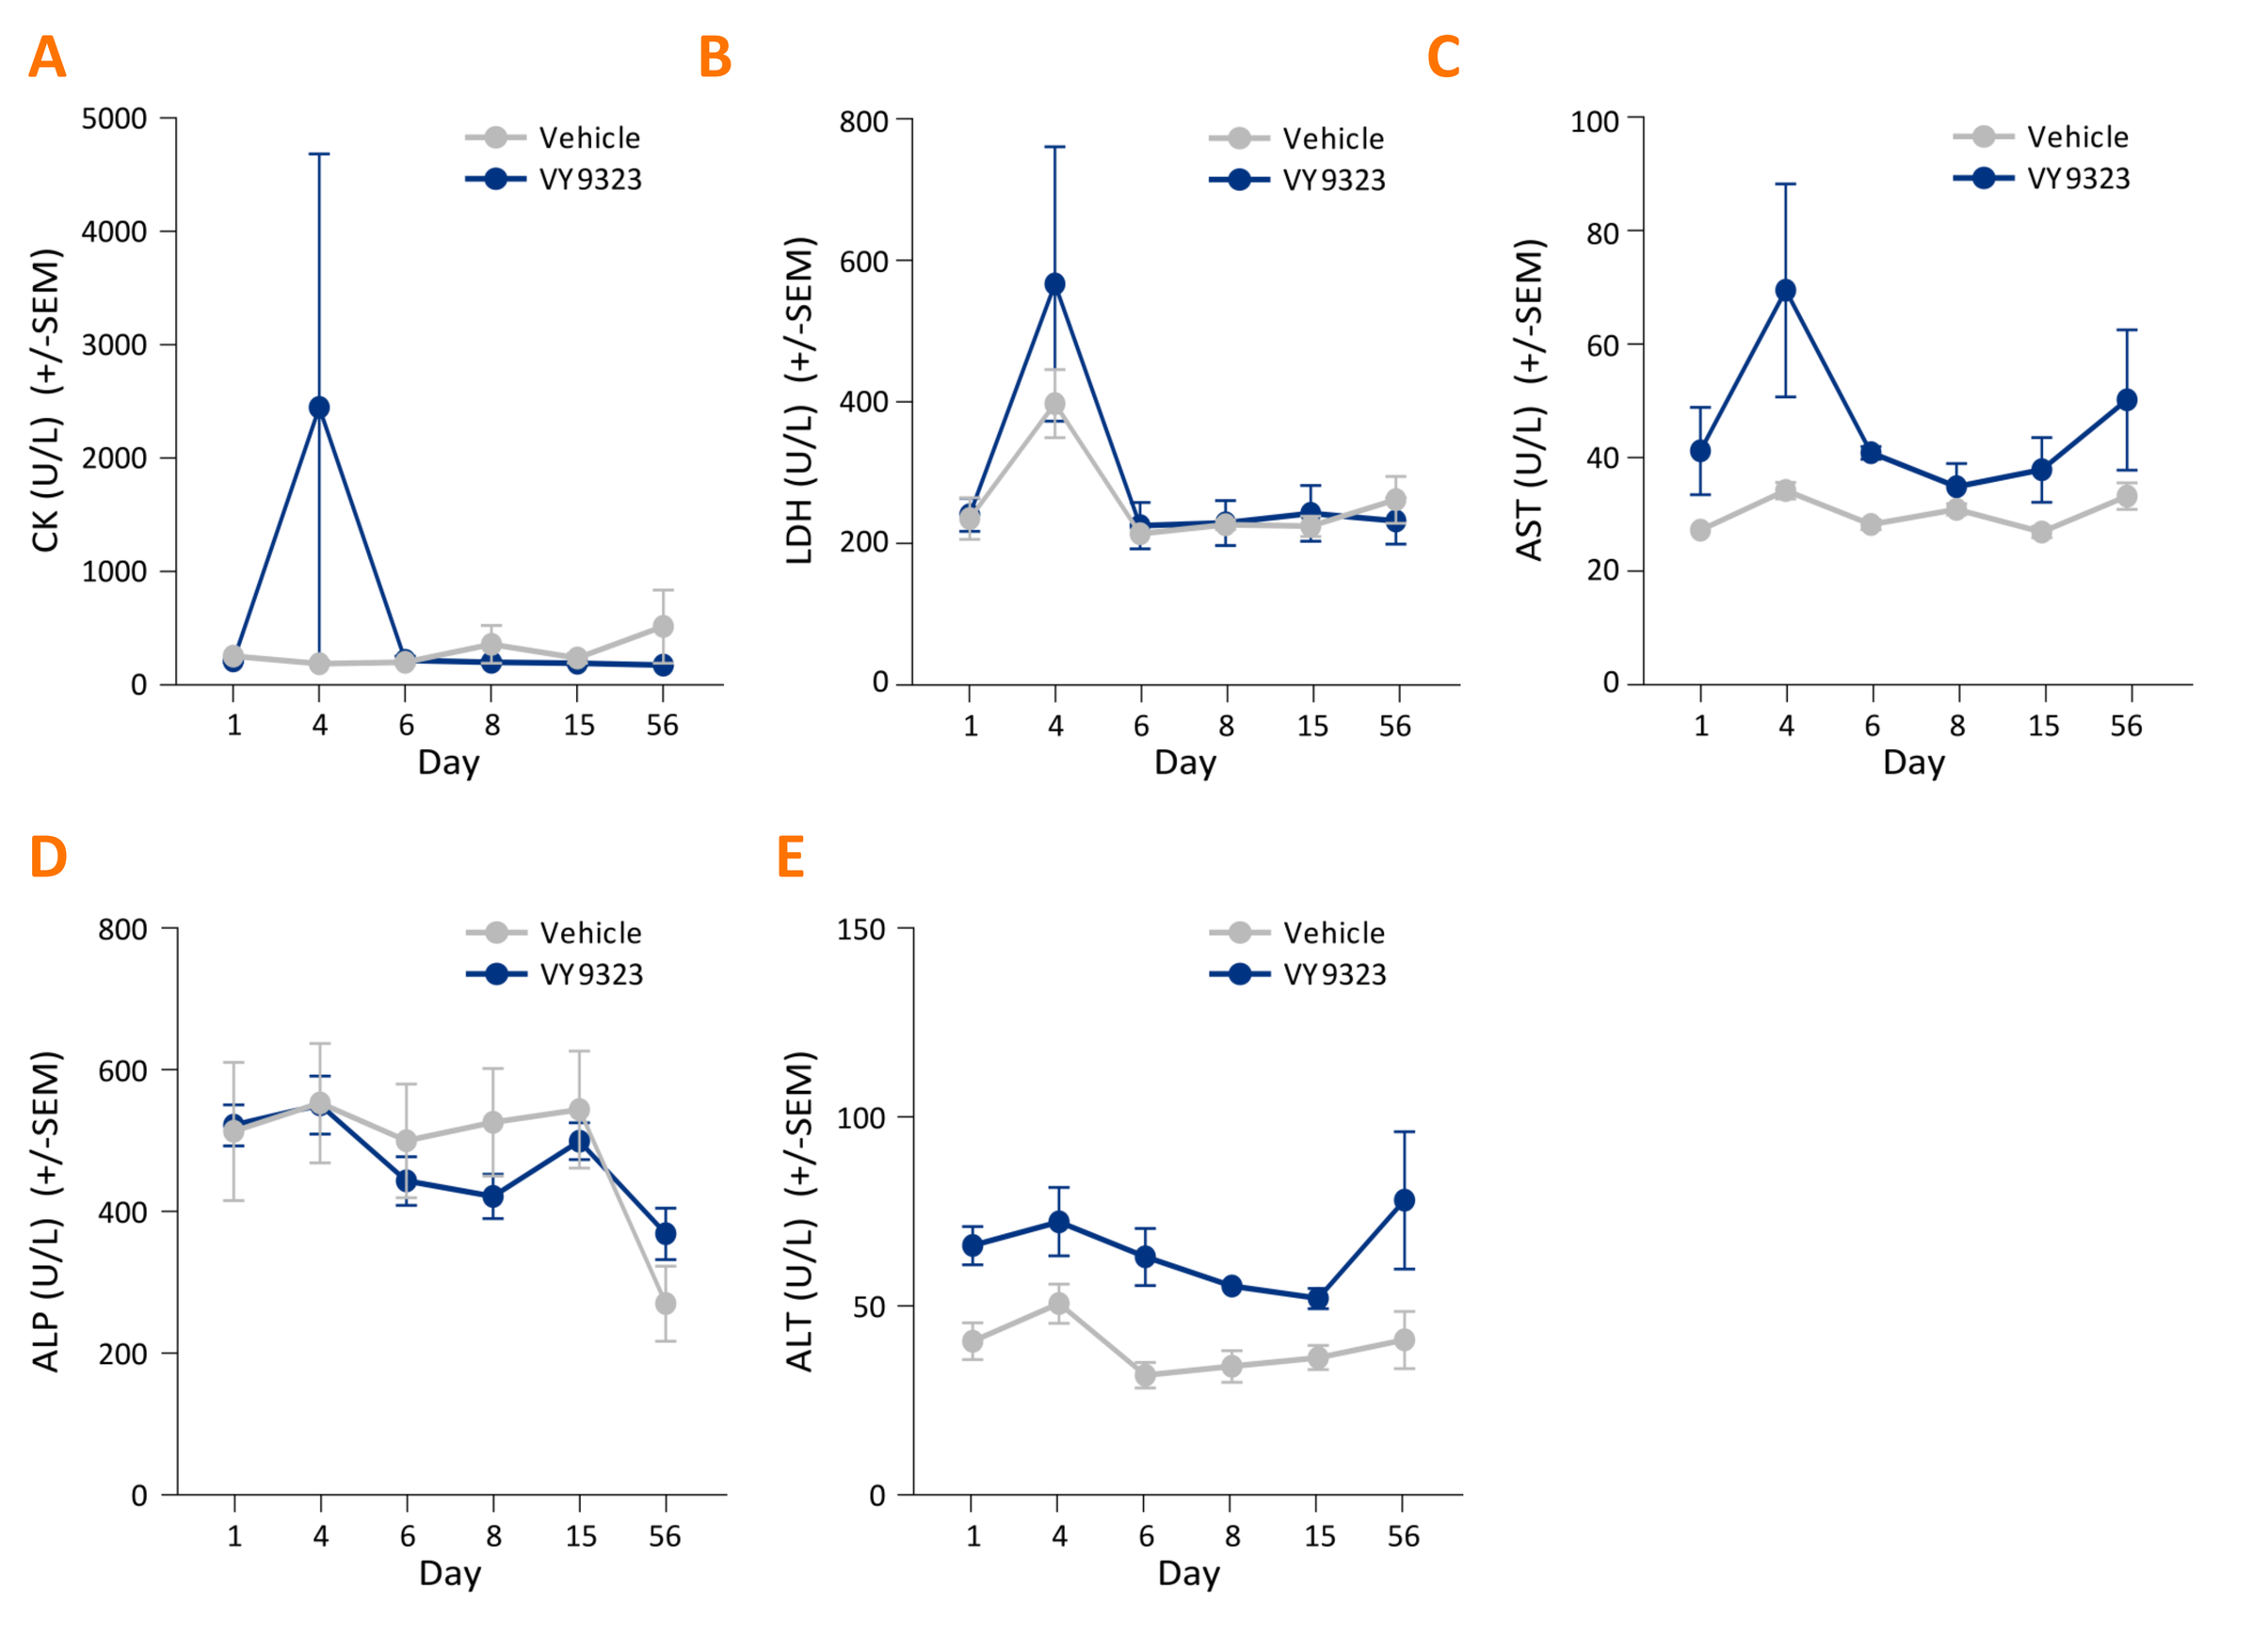

A CK (U/L) (+/-SEM)
B LDH (U/L) (+/-SEM)
C AST (U/L) (+/-SEM)
D ALP (U/L) (+/-SEM)
E ALT (U/L) (+/-SEM)
Day
Vehicle
VY9323

Clinical Chemistry and Hematology findings were uneventful in initial NHP studies. (**A-E**) Moderate transient elevations in ALT, AST, LDH, and CK during first week are typical of AAV administration and are observed following VY9323 administration. (**A**) Further evaluation of CK determined CK-MM (skeletal muscle) was the only isoform elevated in 1 animal.

CONCLUSIONS

- Lead siRNA demonstrates robust potency and selectivity for SOD1 in vitro
- Vectorized miRNA construct produced significant SOD1 reduction and survival benefit in the G93A mouse model of SOD1 ALS
- Identification and characterization of an engineered BBB-penetrant AAV capsid that utilizes a highly conserved cell surface receptor allows for IV delivery and evaluation in NHP
- In primates, robust knockdown of SOD1 in all levels of the spinal cord and motor cortex is observed following IV administration of VY9323, while exhibiting a favorable profile in peripheral tissues
- Motor neuron transduction was demonstrated in Betz cells of the cortex and cholinergic neurons of the hypoglossal nucleus, regions important for ALS disease progression
- These data support the continued development of IV delivered RNAi using a novel BBB-penetrant capsid for use in the clinic

voyager therapeutics 75 Hayden Avenue, Lexington, MA 02421 | www.voyagertherapeutics.com

2024 Annual ALS/MND Meeting | December 6-8, 2024 | Montreal, Canada